A Phase 1 and Pharmacologic Study of MM-111, a Bispecific HER2/HER3 Antibody Fusion Protein, in Combination with Multiple Treatment Regimens in Patients with Advanced HER2 Positive Solid Tumors

D. Richards[1], F. Braiteh[2], S. Anthony[3], J. Edenfield[4], B. Hellerstedt[5], R. Raju[6], P. Conkling[7], C. McDonagh[8], S. Frye[8], V. Moyo[8].

[1]Texas Oncology, Tyler, TX, [2]Comprehensive Cancer Centers of Nevada, Las Vegas, NV, [3]Evergreen Hematology and Oncology, Spokane, WA, [4]Institute of Translational Oncology Research, Greenville, SC, [5]Texas Oncology, Austin, TX, [6]US Oncology, Dayton, OH, [7]Virginia Oncology Associates, Norfolk, VA, [8]Merrimack Pharmaceuticals, Cambridge, MA.

Updated Abstract

•BACKGROUND: Ligand-activated HER3 forms a potent signaling heterodimer with HER2 and is emerging as a key tumorigenic node and mediator of drug resistance. MM-111 (111) is a novel molecule that inhibits ligand activated HER3 signaling in HER2+ tumors. Preclinically, MM-111 potentiates the antitumor activity of and mitigates resistance to trastuzumab, lapatinib and chemotherapies. This study evaluates the safety of MM-111 combined with standard of care (SOC) HER2-targeting regimens, namely; capecitabine, cisplatin, and trastuzumab (Arm 1); lapatinib +/- trastuzumab (Arm 2); and paclitaxel with trastuzumab (Arm 3).

•METHODS: This was a multi-arm Phase 1, dose escalation study of MM-111 in combination with SOC regimens to evaluate safety, pharmacokinetics (PK), and anti-tumor activity. Patients were required to have documented advanced HER2+ cancer, with adequate organ function. Each arm was designed to run as a separate Phase 1 study to address safety and tolerability and each arm utilized a “3 + 3” design with standard dosing of the SOC regimen. MM-111 was dosed weekly at 10 mg/kg and escalated up to 20 mg/kg where possible.

•RESULTS: As of 16 August 2012, 46 patients had been enrolled across three arms.

Study Objectives

Primary:

- To determine the maximum tolerated dose (MTD) and any DLT of MM-111 when administered in combination in patients with HER2+ solid tumors

Secondary:

- To characterize the PK of MM-111 when administered in combination
- To establish the recommended Phase 2 dose(s) of MM-111 when administered in each of the combinations
- To characterize the safety and tolerability of MM-111 when administered in each of the combinations
- To assess the antitumor activity of each combination

Study Schema

Patients with advanced HER2+ solid tumors

Baseline disease evaluation and study eligibility, patient goes on most appropriate treatment

Arm 1
Trastuzumab (2 mg/kg) + cisplatin (80 mg) + capecitabine (1000-800 mg)+ MM-111 (10-5 mg/kg)

Arm 2
Lapatinib (1000 mg) ± trastuzumab (2 mg/kg) + MM-111 (10-20 mg/kg)

Arm 3
Trastuzumab (2 mg/kg) + paclitaxel (80 mg) + MM-111 (10-20 mg/kg)

Determination of the MTD and any DLT of MM-111 in combination, 3+3 design, 3-6 pts/cohort

Patient Demographics

	All n=46	Breast n=29	Other n=17
Age, mean (SD)	60.1 (11.9)	57.3 (10.1)	64.9 (13.4)
Years since diagnosis, median (range)	3.3 (0.6 - 23)	4.6 (0.7 - 23)	1.9 (0.6 - 11)
Female, n (%)	37 (80)	29 (100)	8 (47)
Caucasian, n (%)	41 (89)	25 (86)	16 (94)
Median prior therapies, n	5.1	6.0	3.4

% of Tumor Types

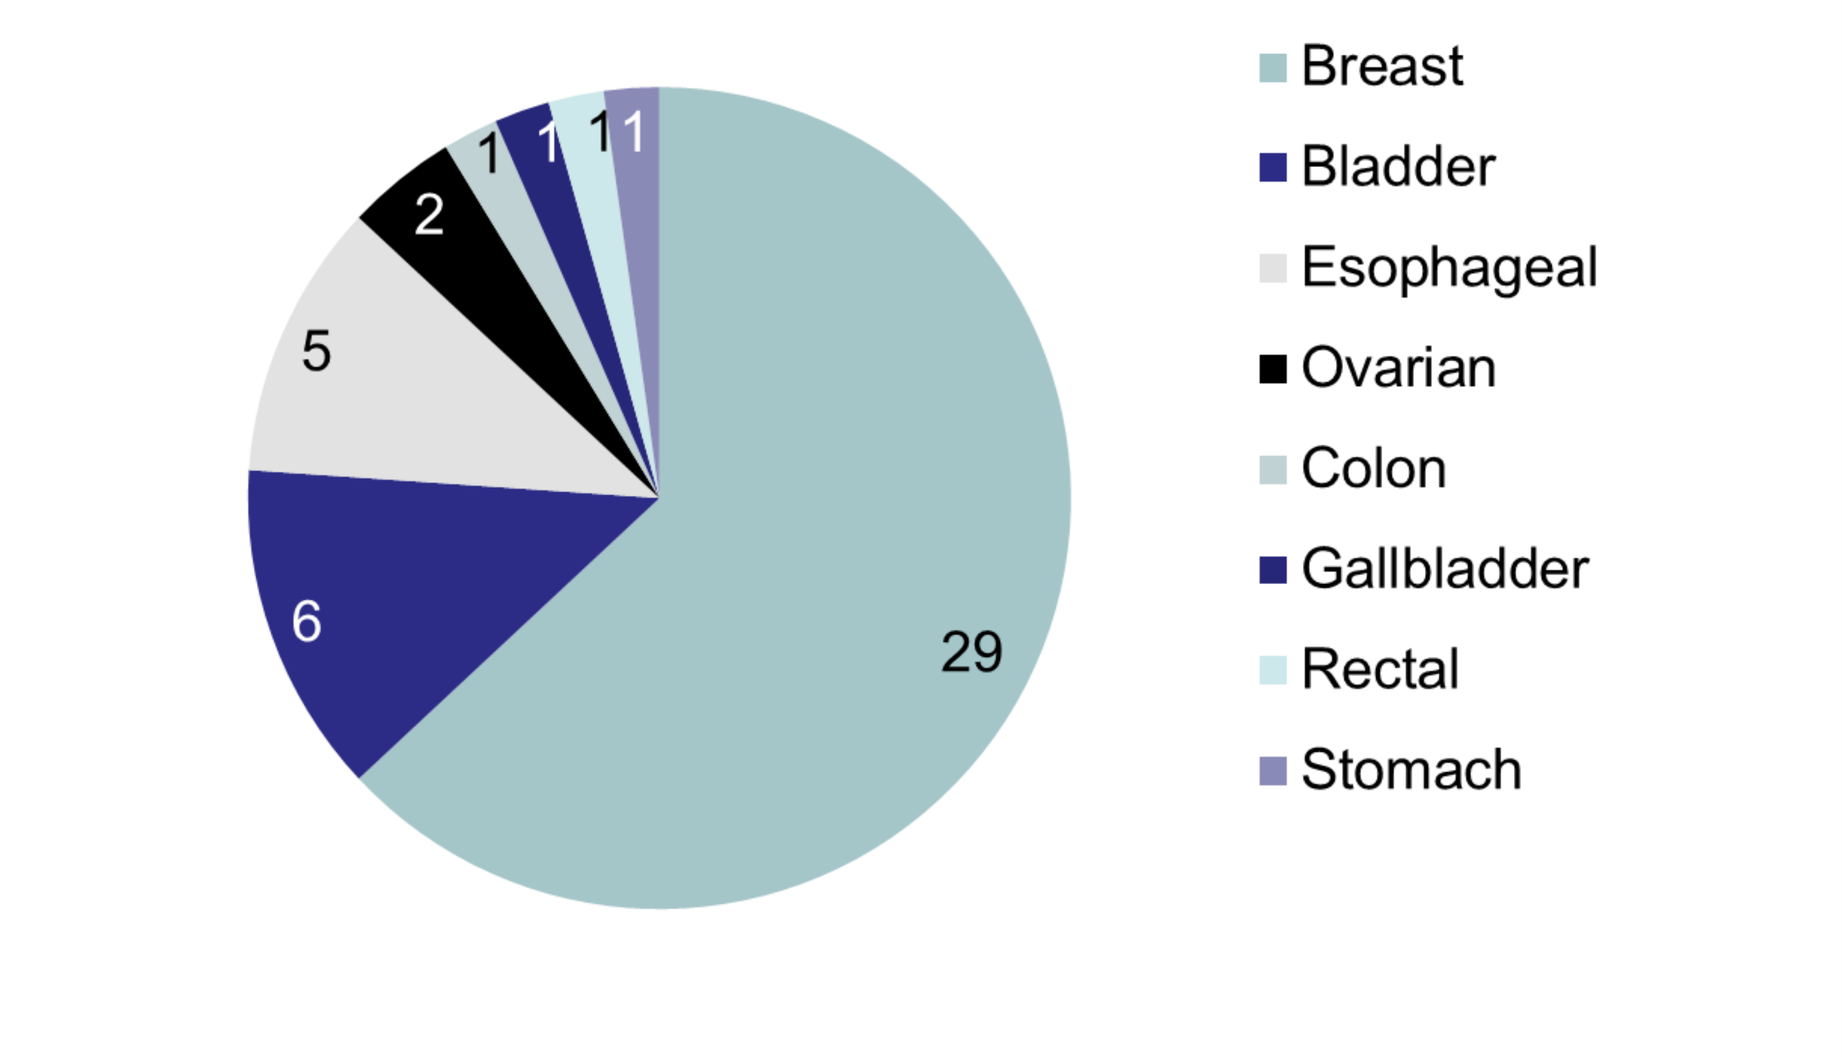

29
6
5
2
1
1
1
1
Breast
Bladder
Esophageal
Ovarian
Colon
Gallbladder
Rectal
Stomach

MM-111

Structure

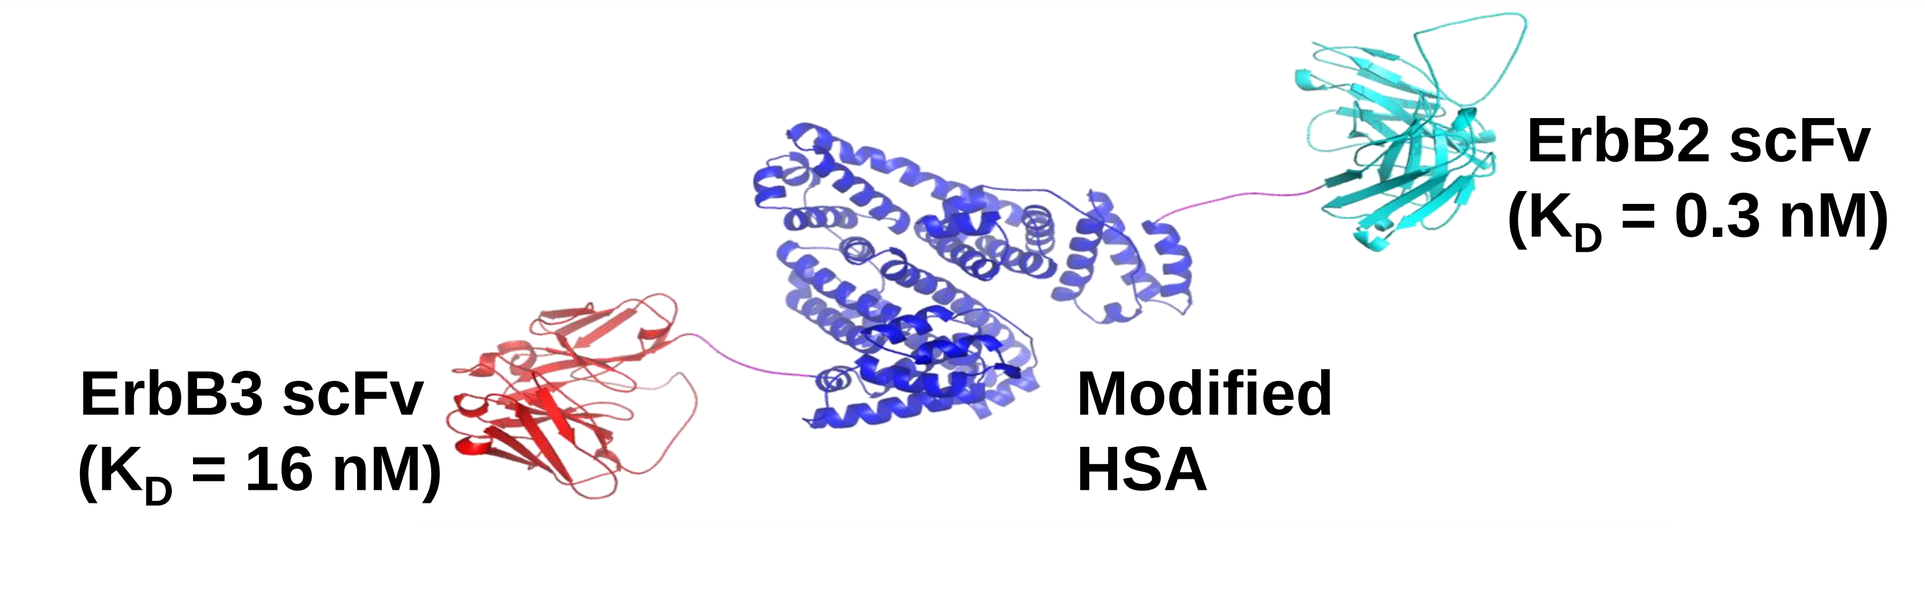

ErbB2 scFv
(K_D = 0.3 nM)
ErbB3 scFv
(K_D = 16 nM)
Modified
HSA

❖HER3 is a key tumorigenic node in HER2-expressing tumors

❖MM-111 is a bi-specific antibody designed to anchor to HER2 and block HER3 activation to treat HER2/HER3-driven disease

Mechanism

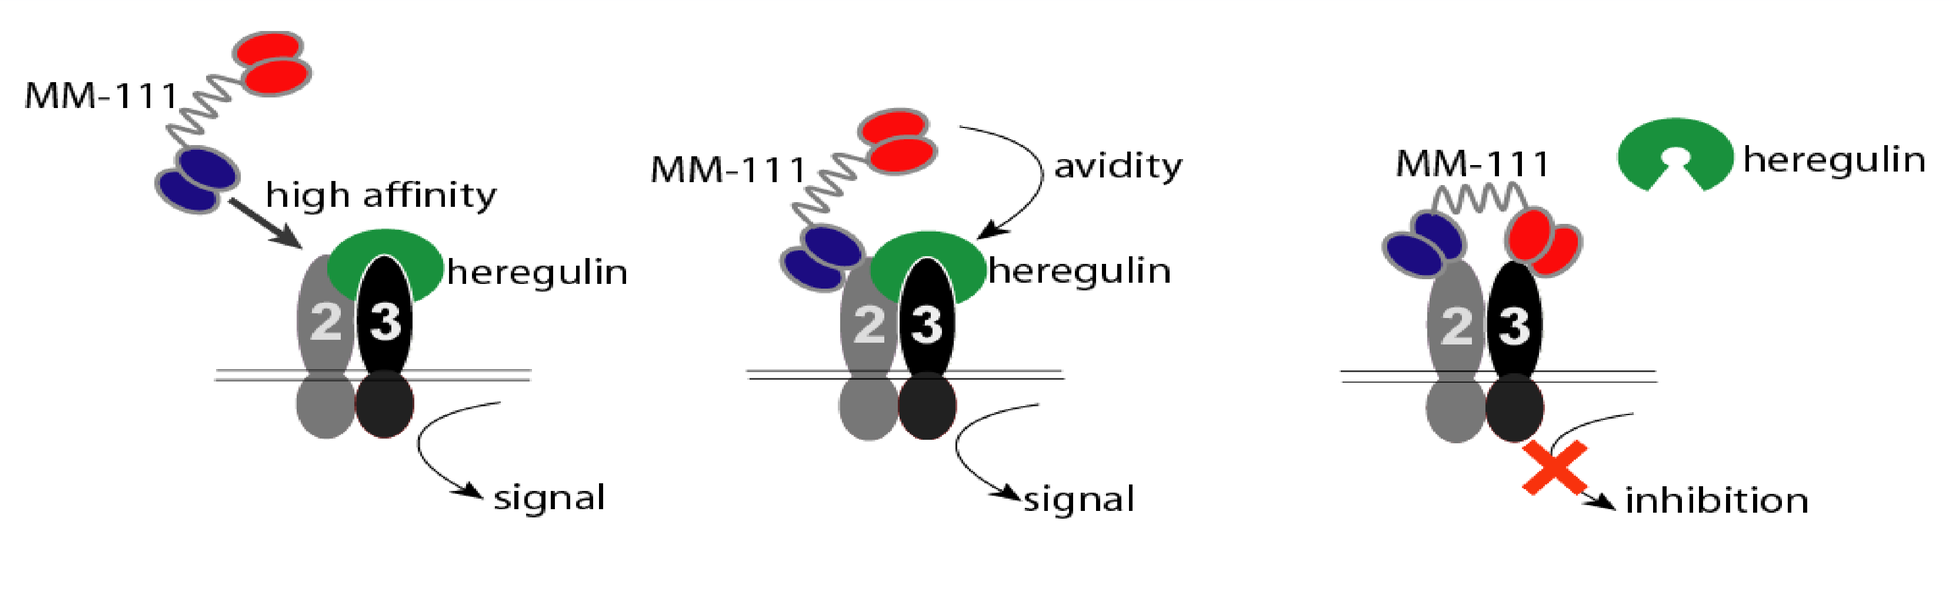

MM-111
high affinity
heregulin
signal
MM-111
avidity
heregulin
signal
MM-111
heregulin
inhibition

❖ Tumors develop resistance to therapy by activating the HER3 signaling axis

❖MM-111 is designed to synergize with a range of therapies by fighting HER3-driven tumor growth and preventing development of resistance

Summary of Safety of MM-111

Adverse Events >20% by Treatment Assignment

System Organ Class Preferred Term	All n = 46 n(%)	Arm1 n = 10 n(%)	Arm2 n = 21 n(%)	Arm3 n = 15 n(%)
GASTROINTESTINAL DISORDERS	40 (87.0)	9 (90.0)	20 (95.2)	11 (73.3)
Diarrhoea	34 (73.9)	8 (80.0)	20 (95.2)	6 (40.0)
Nausea	25 (54.3)	8 (80.0)	12 (57.1)	5 (33.3)
Vomiting	12 (26.1)	6 (60.0)	4 (19.0)	2 (13.3)
GENERAL DISORDERS AND ADMINISTRATION SITE CONDITIONS	35 (76.1)	10 (100)	16 (76.2)	9 (60.0)
Fatigue	26 (56.5)	10 (100)	10 (47.6)	6 (40.0)
METABOLISM AND NUTRITION DISORDERS	34 (73.9)	10 (100)	14 (66.7)	10 (66.7)
Decreased appetite	18 (39.1)	8 (80.0)	5 (23.8)	5 (33.3)
Dehydration	10 (21.7)	7 (70.0)	3 (14.3)	
Hypokalaemia	20 (43.5)	8 (80.0)	8 (38.1)	4 (26.7)
Hypomagnesaemia	12 (26.1)	8 (80.0)	2 (9.5)	2 (13.3)
SKIN AND SUBCUTANEOUS TISSUE DISORDERS	24 (52.2)	5 (50.0)	11 (52.4)	8 (53.3)
RESPIRATORY, THORACIC AND MEDIASTINAL DISORDERS	22 (47.8)	6 (60.0)	10 (47.6)	6 (40.0)
Dyspnoea	12 (26.1)	3 (30.0)	6 (28.6)	3 (20.0)
BLOOD AND LYMPHATIC SYSTEM DISORDERS	21 (45.7)	9 (90.0)	3 (14.3)	9 (60.0)
Anaemia	18 (39.1)	8 (80.0)	1 (4.8)	9 (60.0)
Neutropenia	12 (26.1)	7 (70.0)	1 (4.8)	4 (26.7)
NERVOUS SYSTEM DISORDERS	21 (45.7)	5 (50.0)	10 (47.6)	6 (40.0)
MUSCULOSKELETAL AND CONNECTIVE TISSUE DISORDERS	19 (41.3)	4 (40.0)	7 (33.3)	8 (53.3)
INVESTIGATIONS	18 (39.1)	3 (30.0)	12 (57.1)	3 (20.0)
PSYCHIATRIC DISORDERS	17 (37.0)	6 (60.0)	5 (23.8)	6 (40.0)
INFECTIONS AND INFESTATIONS	16 (34.8)	4 (40.0)	6 (28.6)	6 (40.0)

Dose Limiting Toxicities (DLT) by Treatment Arm*

Arm 1 Cohort 1 (10 mg/kg)

Subject ID	MedDRA Preferred Term	CTCAE
38-11-021	Diarrhoea	Grade 3
40-11-022	Diarrhoea	Grade 3
	Febrile neutropenia	Grade 3
	Mucosal inflammation	Grade 3
	Nausea	Grade 3
	Vomiting	Grade 3
37-11-010	Hyperuricaemia	Grade 4

Arm 1 Cohort 2 (5 mg/kg)

Subject ID	MedDRA Preferred Term	CTCAE
44-12-027	Hypokalaemia	Grade 3
	Hyponatraemia	Grade 3
	Neutropenia	Grade 3
45-12-030	Neutropenia	Grade 4
	Anaemia	Grade 3
	Thrombocytopenia	Grade 3

Arm 2 Cohort 2 (20 mg/kg)

Subject ID	MedDRA Preferred Term	CTCAE
46-22-039	Hypokalaemia	Grade 3

Arm 3 Cohort 1 (10 mg/kg)

Subject ID	MedDRA Preferred Term	CTCAE
37-31-001	Hypophosphataemia	Grade 3

* No DLTs were observed in Arm 2 Cohort 1 or Arm 3 Cohort 2

Summary of Cardiac Safety and Pharmacokinetics

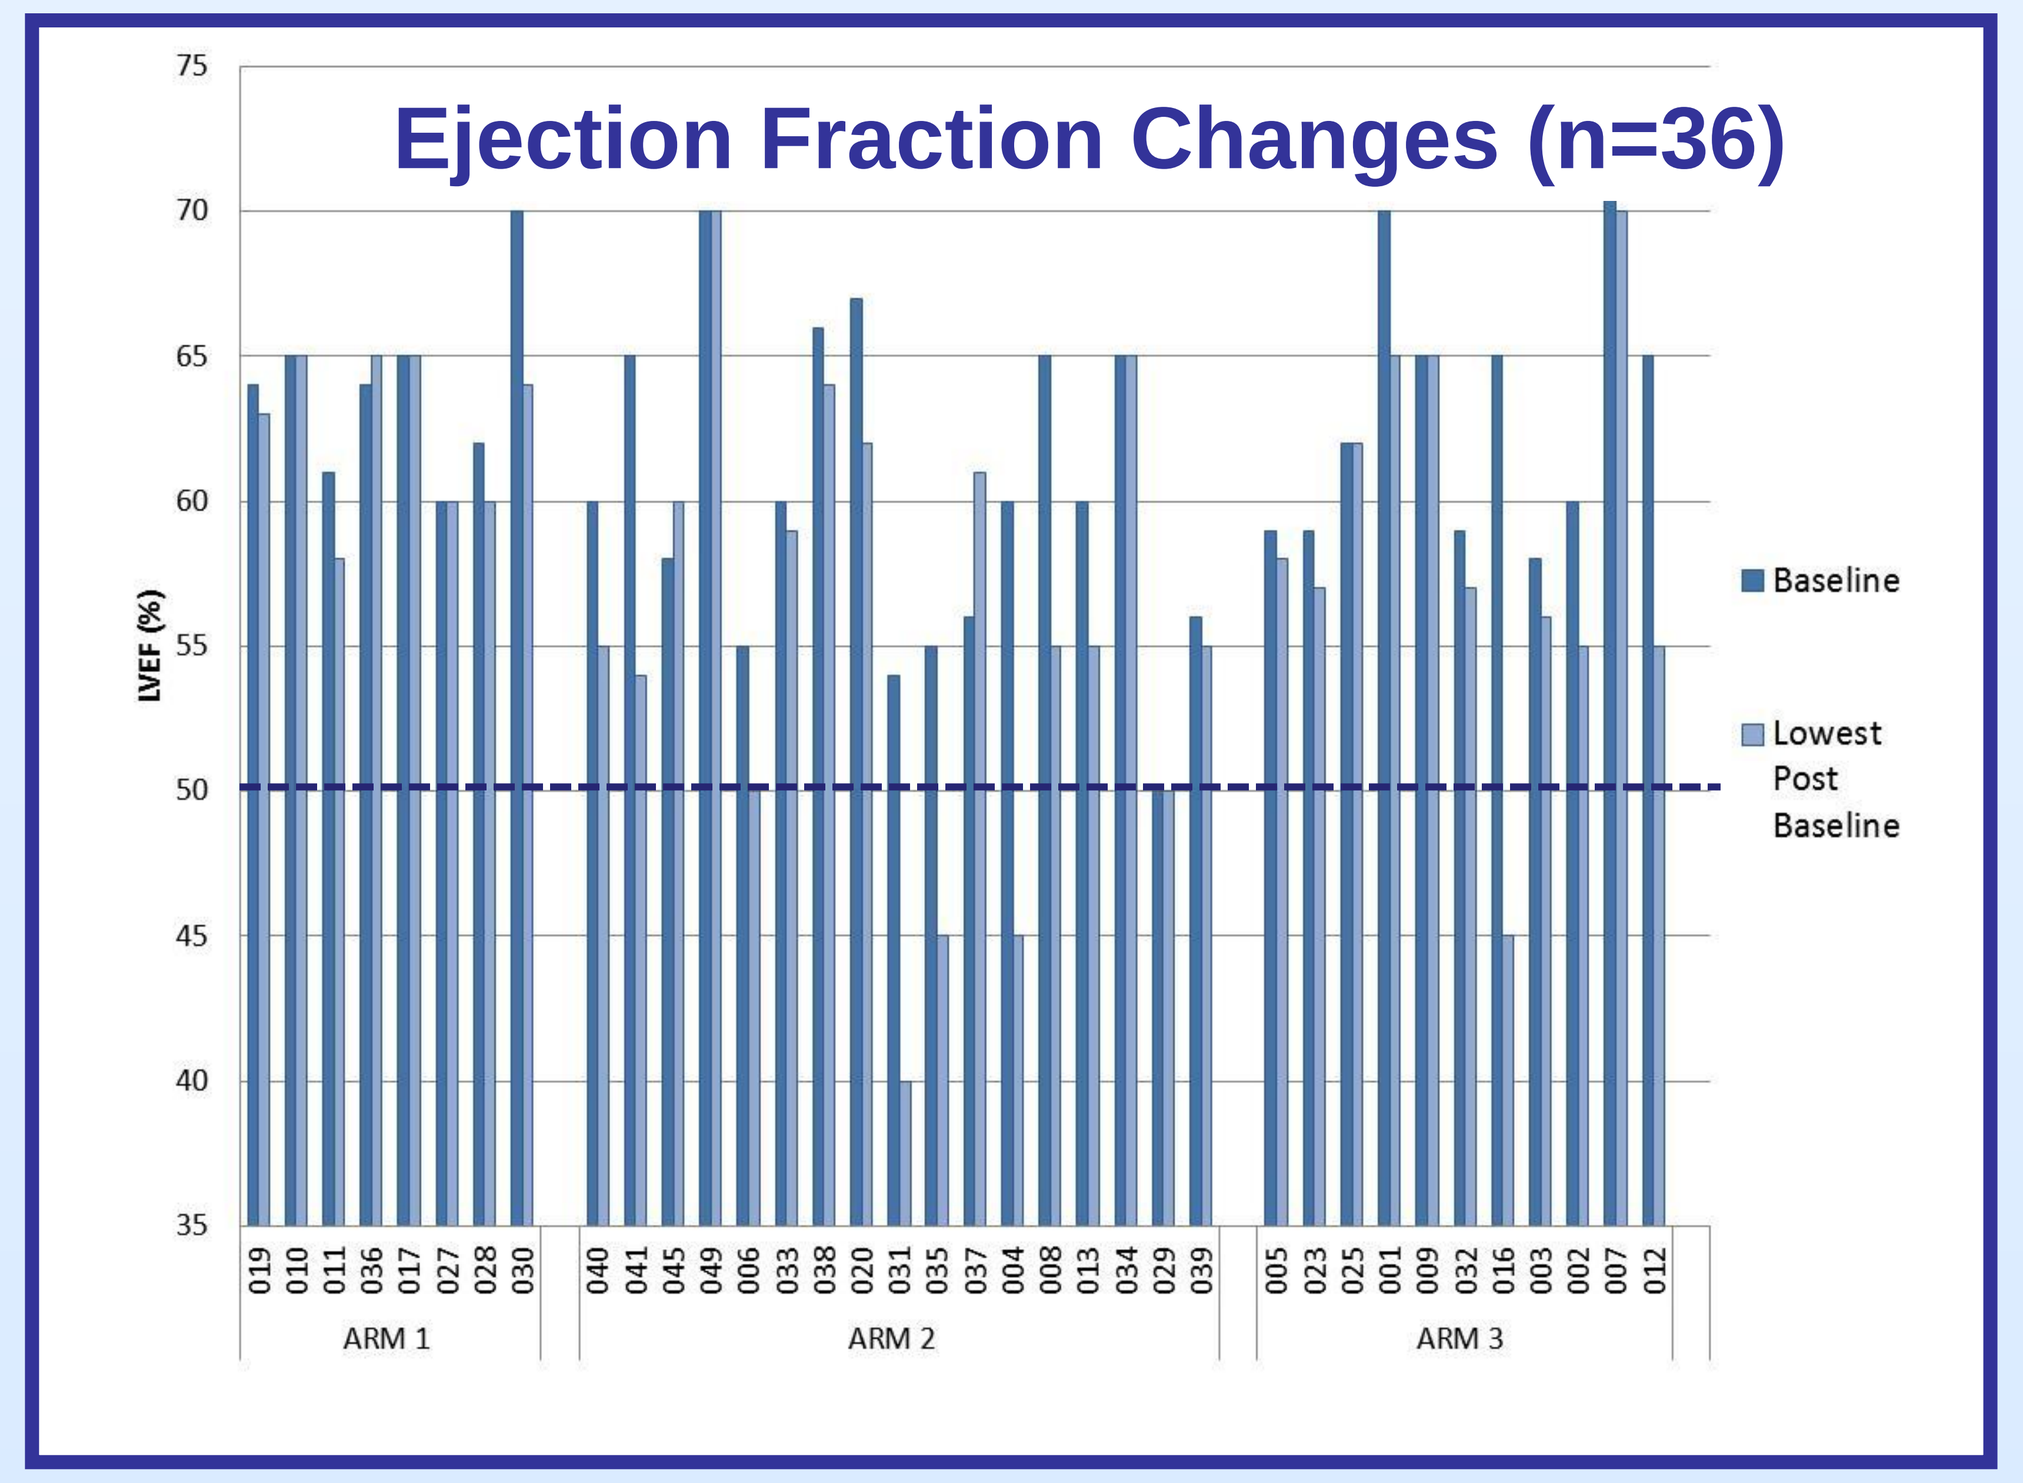

Ejection Fraction Changes (n=36)
LVEF (%)
Baseline
Lowest Post Baseline
ARM 1
ARM 2
ARM 3

LVEF

- ARM2 pt 031 diagnosed with CHF on study with extensive chest wall disease with malignant pleural effusion and history of prior anthracyline exposure

Phamacokinetics

- 10 mg (15 pts): $t_{1/2}$ = 90 ± 26 hr
 Cmax = 192 ± 41 ug/mL
 AUC = 13.7 ± 5330 mg/mL*hr
 CL = 0.69 ± 0.15 mL/kg/hr
- 20 mg (6 pts): $t_{1/2}$ = 86 ± 55 hr
 Cmax = 474 ± 155 ug/mL
 AUC = 32.2 ± 10539 mg/mL*hr
 CL = 0.61 ± 0.14 mL/kg/hr

Summary of Response

Best Response for Target Lesions by Patient

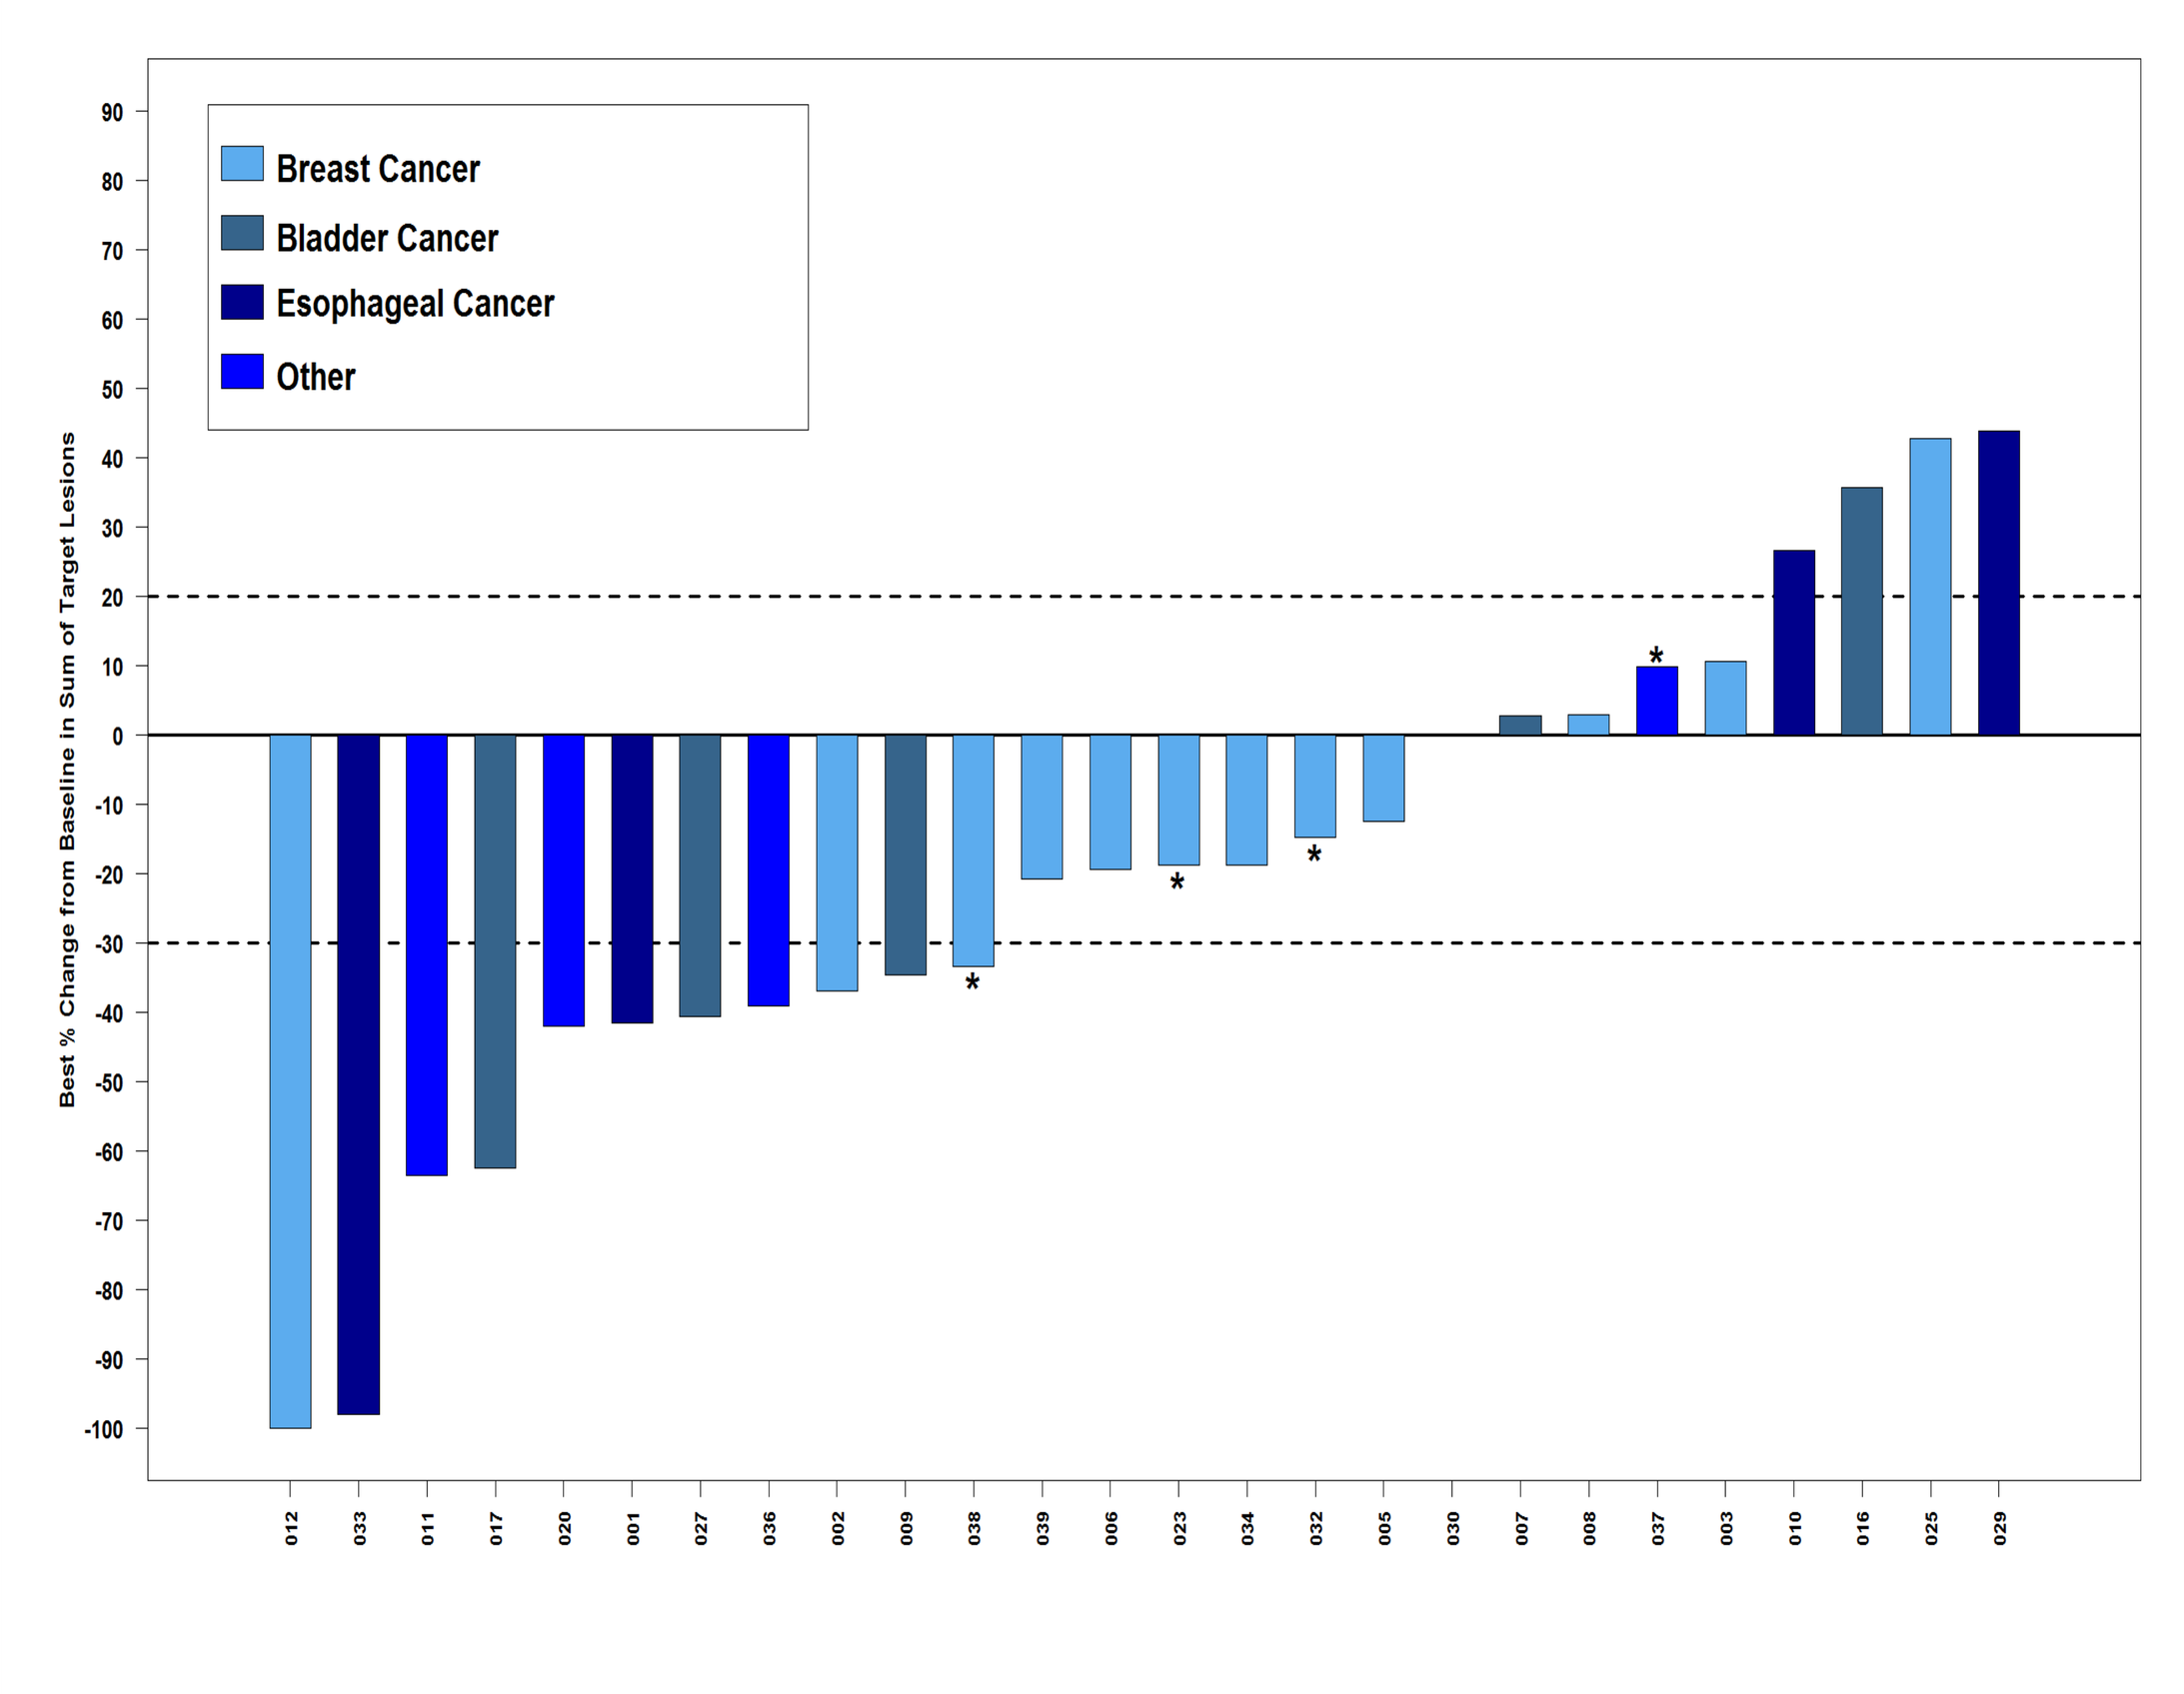

Breast Cancer
Bladder Cancer
Esophageal Cancer
Other
Best % Change from Baseline in Sum of Target Lesions

* Indicates patient ongoing

Response by Treatment Arm

Treatment	n	Not Evaluable: died	Not Evaluable: sympt det	Not Evaluable: other term.	Not Evaluable: No f/u scan yet	Evaluable: CR	Evaluable: PR	Evaluable: SD	Evaluable: PD
Arm 1	**10**			3		1	4	1	1
Arm 2	**21**	2	2	2	3		3	5	4
Arm 3	**15**				4		4	4	3
Total	**46**	2	2	5	7	1	11	10	8

Response by Tumor Type

Cancer Type	n (%)	Not Evaluable: died	Not Evaluable: symp det	Not Evaluable: other term.	Not Evaluable: No f/u scan yet	Evaluable: CR	Evaluable: PR	Evaluable: SD	Evaluable: PD
Breast	20 (63)	1 (3)	2 (7)	4 (14)	6 (21)	1 (3)	3 (10)	9 (31)	3 (10)
Bladder	5 (13)			1 (17)			2 (50)		2 (33)
Esoph-ageal	5 (11)				1 (20)		2 (40)		2 (40)
Other	7 (13)	1 (17)					4 (50)	1 (17)	1 (17)
Total	46 (100)	2 (4)	2 (40)	5 (11)	7 (15)	1 (2)	11 (24)	10 (22)	8 (17)

Conclusions

- MM-111 can be safely combined with various HER2 targeting regimens
 - Dose escalation of MM-111 at 20 mg/kg is feasible with both lapatinib/trastuzumab and paclitaxel/trastuzumab
 - Most DLTs were observed in the Arm 1 regimen and were attributable to capecitabine
 - The capecitabine dose in Arm 1 has been de-escalated to 800 mg/m2 BID and re-escalation of MM-111 is ongoing
- The safety profile of the MM-111 combinations are consistent with that of the underlying HER2 therapy backbone
- A clinical benefit rate (CR, PR, SD > 4 months) of 52% in 29 evaluable patients was observed across all treatment arms
- Future studies will evaluate the activity of MM-111 in several HER2-expressing tumor types

Contacts

- Dr. Donald Richards: donald.richards@usoncology.com
- Dr. Victor Moyo: vmoyo@merrimackpharma.com